Development and validation of baseline predictive biomarkers for response to immuno-checkpoint treatments in the context of multiline and multitherapy cohorts using EpiSwitch™ epigenetic profiling

E. Hunter[1], S. Potluri[2], S. Zhang[3], M. Dezfouli[1], J. Back[1], L. James[1], N. Jandor[1], R. Powell[1], M. Salter[1], A. Ramadass[1], J. Green[1], W. Westra[1], H. Dong[4], R. Dronca[4], S. N. Markovic[4], P. B. Robbins[5], T. Cai[3], A. Akoulitchev[1], and P. K. Shah[3]

[1]Oxford Biodynamics, Oxford, UK; [2]Lyell Immunopharma, San Francisco, CA, USA; [3]EMD Serono Research & Development Institute, Billerica, MA, USA; [4]Mayo Clinic, Rochester, MN, USA; [5]Pfizer, La Jolla, CA, USA

INTRODUCTION

- The EpiSwitch biomarker discovery platform detects systemic changes in the cellular genomic architecture using a microarray and PCR-based biomarker platform (**Figure 1**).[1] It identifies and monitors chromosome conformation signatures (CCSs), key regulatory processes that integrate environmental cues and genetic alterations into the genomic regulatory machinery
 - CCSs are also known as gene loops, chromosome domains, or long-range chromosomal interactions
- CCSs are the primary step in a cascade of genomic regulatory events and are directly linked with cellular phenotype. CCSs have low off-rates and only change when a physiologically signaled transition to a new phenotype occurs or because of external intervention. As these events are binary, they can be measured by changes in other cellular entities, such as DNA methylation and RNA and protein expression
- We profiled patients with non-small cell lung cancer (NSCLC) and melanoma treated with avelumab (an anti–PD-L1 antibody), pembrolizumab (an anti–PD-1 antibody), or pembrolizumab in combination with the chemotherapeutic agent azacitidine (**Table 1**) and generated models to differentiate responders from nonresponders using machine learning methods

Figure 1. EpiSwitch biomarker methodology: from discovery to clinical assay

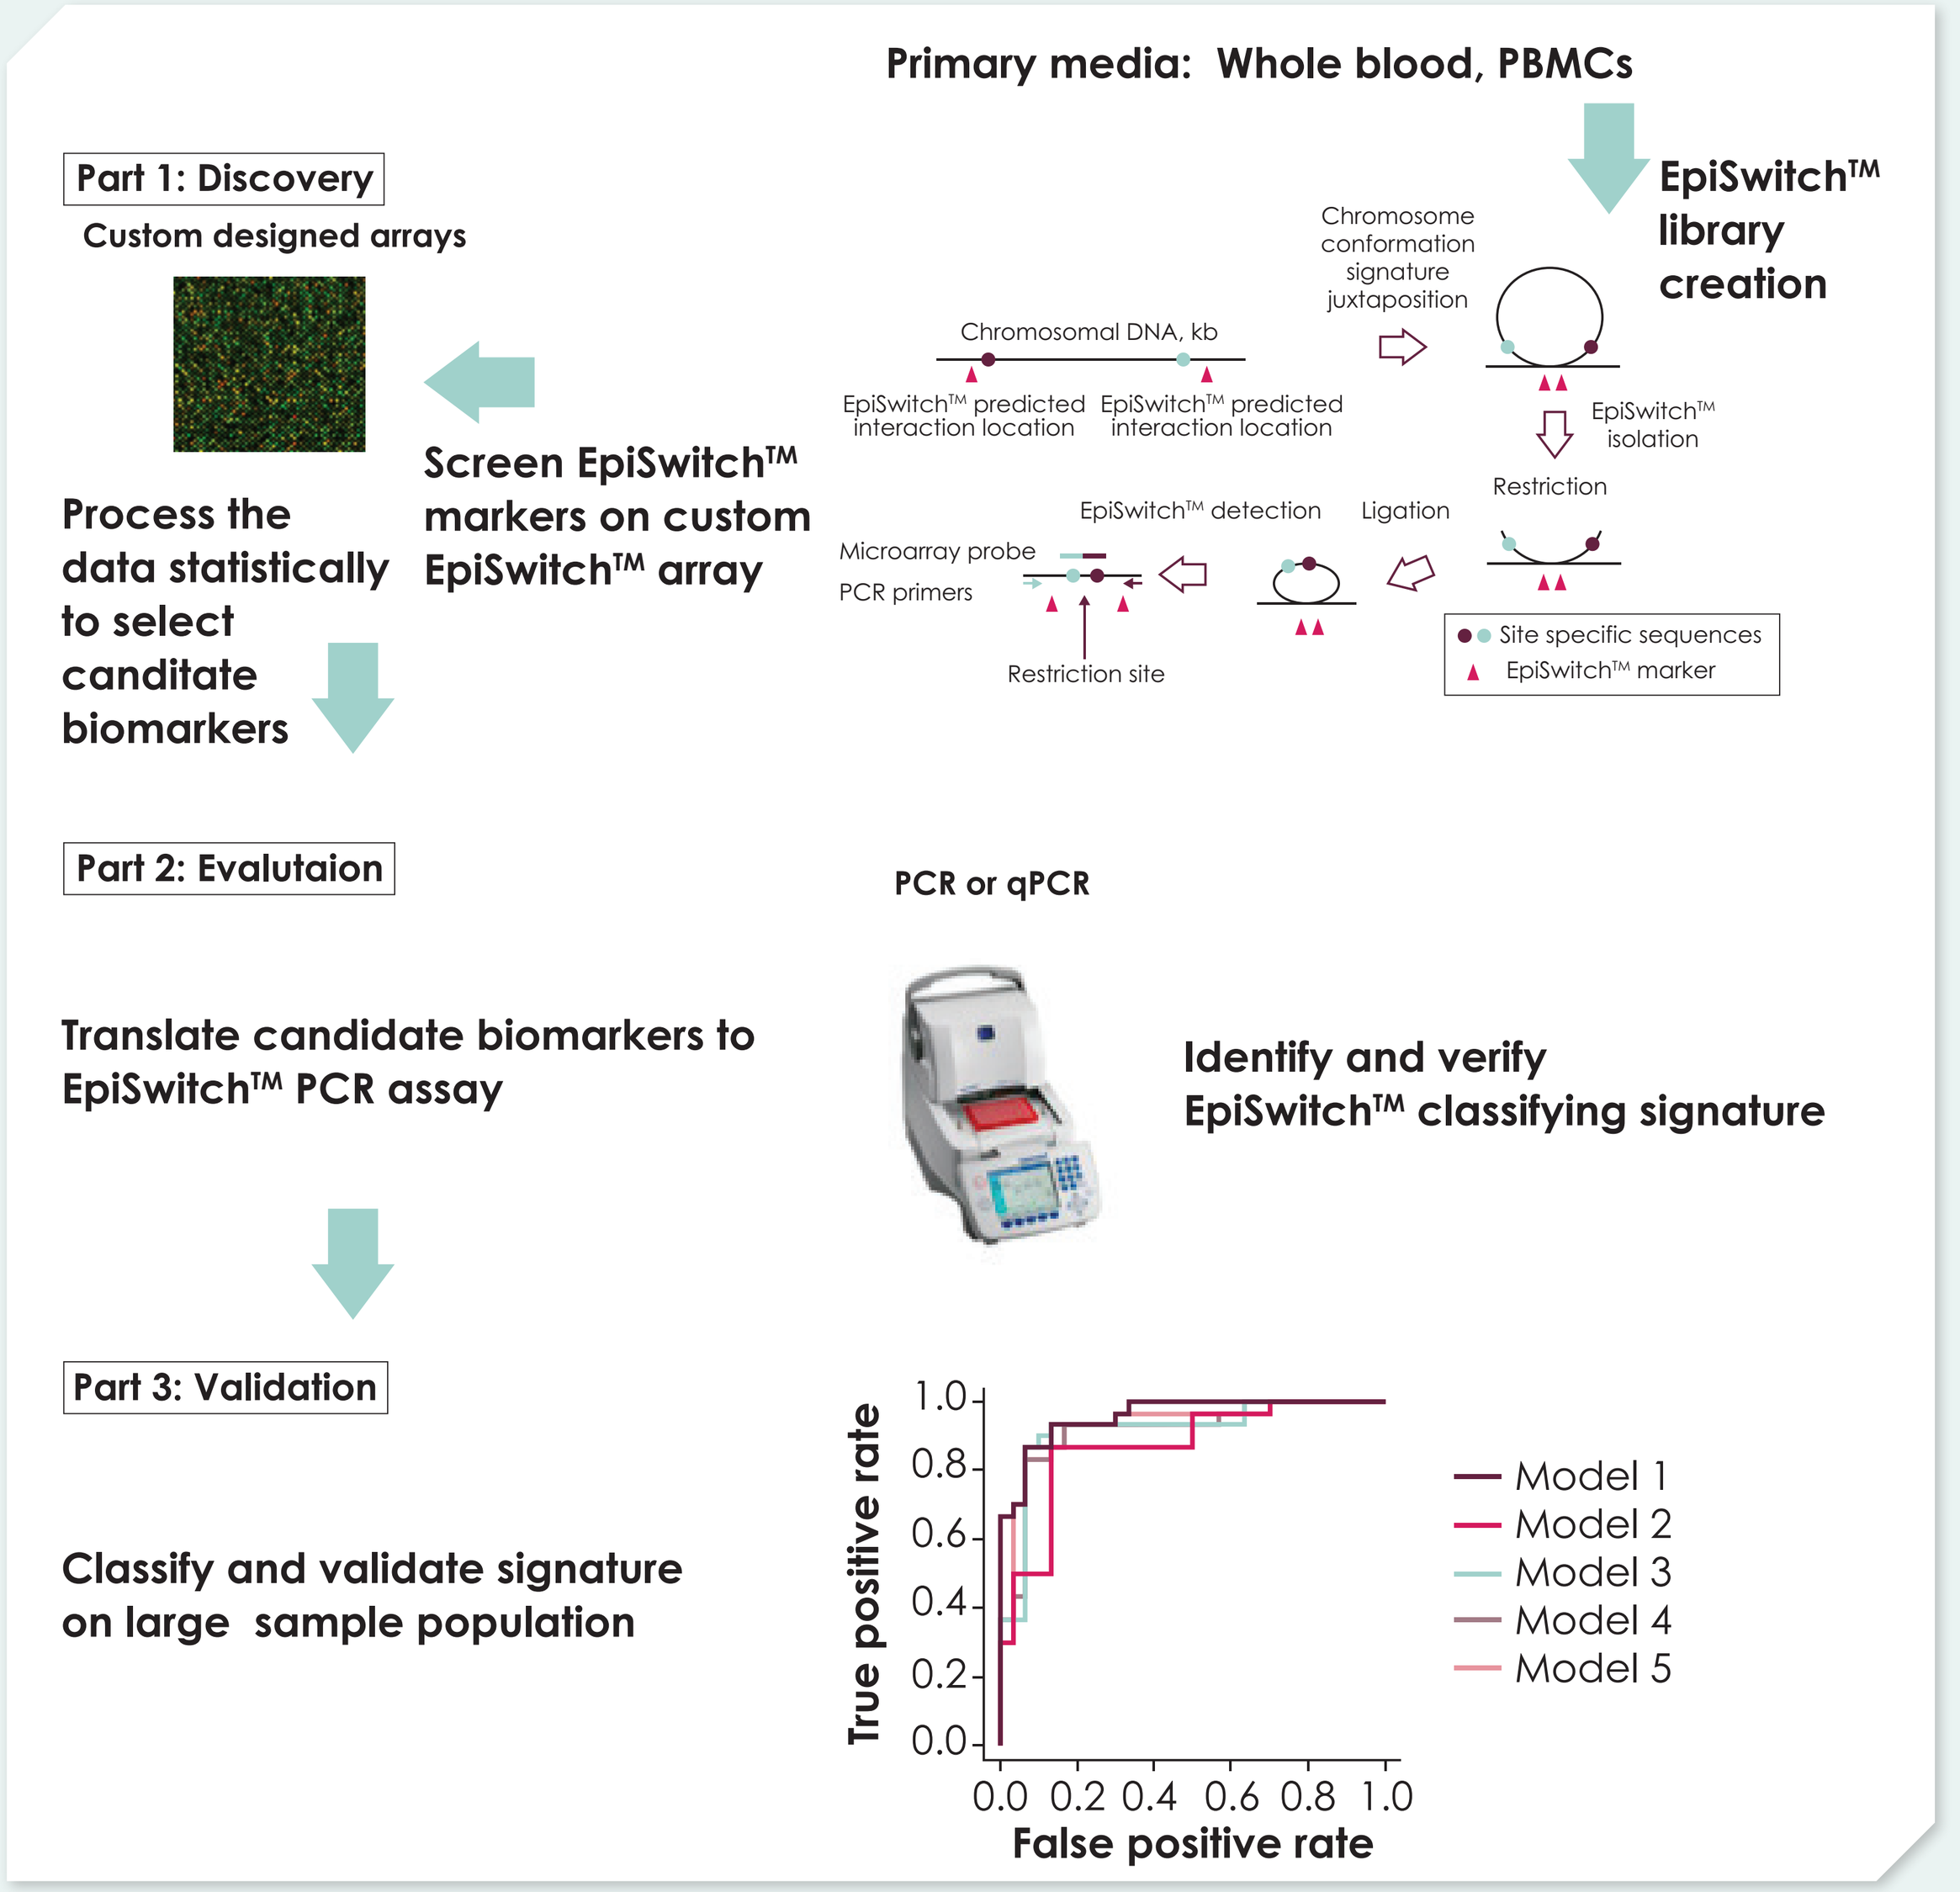
Primary media: Whole blood, PBMCs
Part 1: Discovery
Custom designed arrays
Screen EpiSwitch™ markers on custom EpiSwitch™ array
Process the data statistically to select canditate biomarkers
EpiSwitch™ library creation
Chromosome conformation signature juxtaposition
Chromosomal DNA, kb
EpiSwitch™ predicted interaction location
EpiSwitch™ predicted interaction location
EpiSwitch™ isolation
Restriction
EpiSwitch™ detection
Ligation
Microarray probe
PCR primers
Restriction site
Site specific sequences
EpiSwitch™ marker
Part 2: Evalutaion
PCR or qPCR
Translate candidate biomarkers to EpiSwitch™ PCR assay
Identify and verify EpiSwitch™ classifying signature
Part 3: Validation
Classify and validate signature on large sample population
True positive rate
False positive rate
Model 1
Model 2
Model 3
Model 4
Model 5

Table 1. Data sets analyzed using EpiSwitch

Treatment, n	Disease	CR	PR	SD	PD	NE	Total
		Clinical response per RECIST 1.1					
Avelumab	1L NSCLC	1	17	10	16	6	50
Avelumab	2L NSCLC	2	17	8	21	1	49
Pembrolizumab	2L NSCLC	0	7	12	25	0	44
Pembrolizumab	Melanoma	4	0	0	4	0	8
Pembrolizumab + azacitidine	2L NSCLC	0	9	2	29	0	40

1L, first-line; **2L**, second-line; **CR**, complete response; **NE**, not evaluable; **PD**, progressive disease; **PR**, partial response; **SD**, stable disease

METHODS

- The classifiers were generated in 2 steps using n-fold cross-validation:
 - Step 1: identification of univariate association between biomarkers and response using Fisher exact test
 - Step 2: generation of multivariate model using XGBoost or regularized regression
- A multivariable Cox proportional hazards model was built using R package survival and plotted using Survminer
- For building multivariate models, appropriate clinical covariates were selected based on analysis of clinical trial data from the JAVELIN Lung 200 trial (avelumab vs docetaxel in patients with platinum-treated advanced NSCLC; NCT02395172)[2]
- A multivariable logistic regression model was built with glm function

RESULTS

- The 2L NSCLC classifier, built using baseline profiles of avelumab-treated patients, achieved good generalization performance for predicting response in a 2L NSCLC test set; however, this classifier did not have acceptable classification performance for predicting response in 1L NSCLC (see poster P142)
- This was also the case when the baseline 1L NSCLC classifier was used to predict response in a 2L NSCLC test set (see poster P142)
- However, the composite classifier trained using baseline profiles of both 1L and 2L patients achieved good generalized performance for both treatment lines (**Figure 2**)
- This classifier also had good positive-predictive value (PPV) when identifying responders to pembrolizumab from baseline profiles (**Figure 2**)
 - Pembrolizumab, like avelumab, inhibits the PD-L1/PD-1 pathway
- However, the negative predictive value (NPV) was reduced, suggesting that responders to both avelumab and pembrolizumab may have similar profiles but the profiles of non-responders to these drugs could be different

Figure 2. Generation of indication-agnostic NSCLC classifier

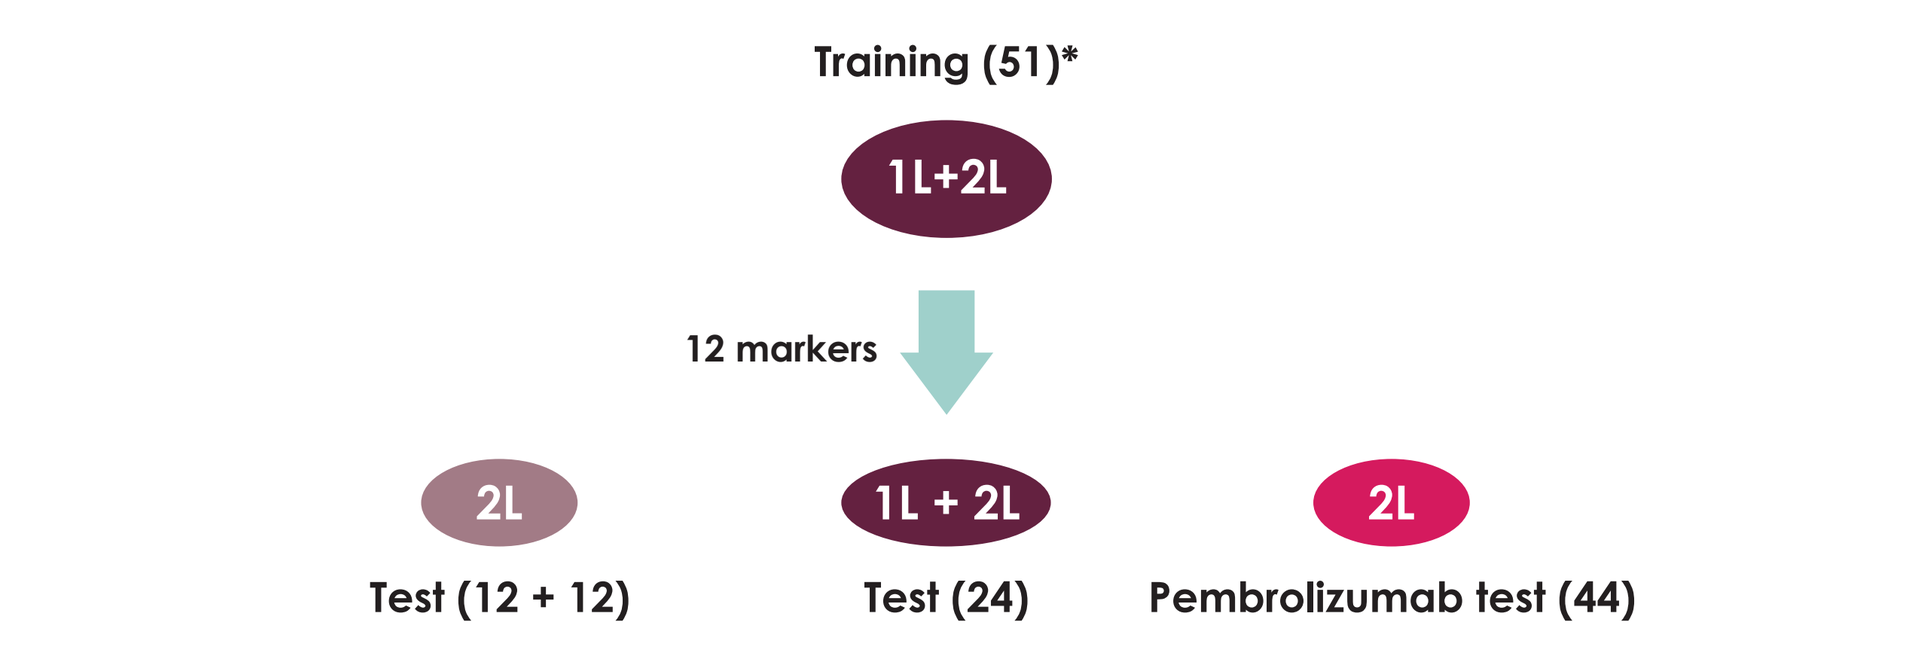
Training (51)*
1L+2L
12 markers
2L
Test (12 + 12)
1L + 2L
Test (24)
2L
Pembrolizumab test (44)

Performance on response

	Training set	2L Test set	1L Test set	Pembrolizumab 2L
Accuracy (95% CI)	0.90 (0.80-0.97)	0.75 (0.53-0.90)	0.76 (0.52-0.92)	0.45 (0.30-0.61)
PPV	0.87	0.75	0.75	**0.74**
NPV	0.96	0.75	0.78	0.24

* Patients with SD were taken as responders and used in training/test

- There was a significant difference (log-rank p<0.001) in progression-free survival (PFS) and overall survival (OS) between the predicted response groups (**Figure 3**)
- This survival difference was not observed with PD-L1 when patients were stratified at different thresholds of PD-L1 positivity (**Figure 4**)

Figure 3. OS plots for classifier predictions

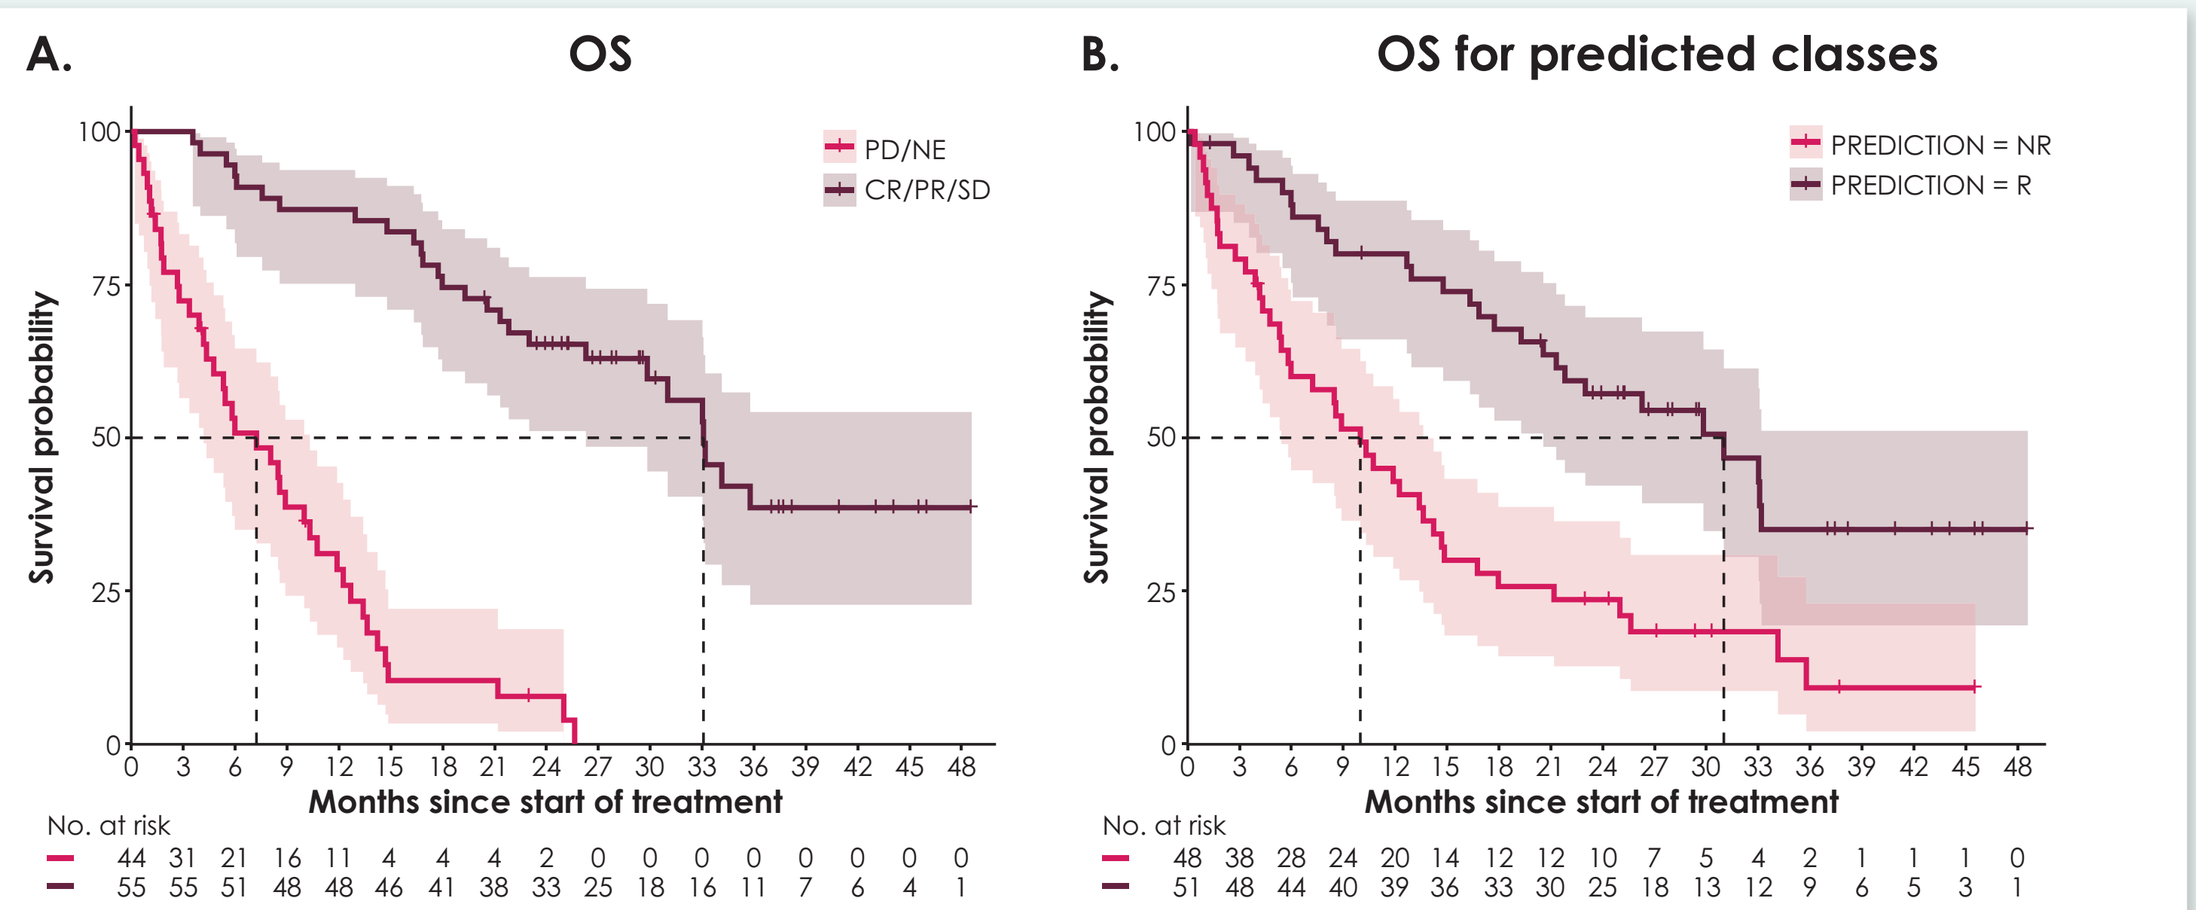
A.
OS
PD/NE
CR/PR/SD
B.
OS for predicted classes
PREDICTION = NR
PREDICTION = R
Survival probability
Months since start of treatment
No. at risk

A.

Response	n	Events	Median	0.95 LCI	0.95 UCI
NR (PD/NE)	44	40	7.23	4.17	10.32
R (CR/PR/SD)	55	27	33.08	26.28	NA

B.

Response	n	Events	Median	0.95 LCI	0.95 UCI
NR (PD/NE)	48	40	9.99	5.42	13.63
R (CR/PR/SD)	51	27	31.01	20.57	NA

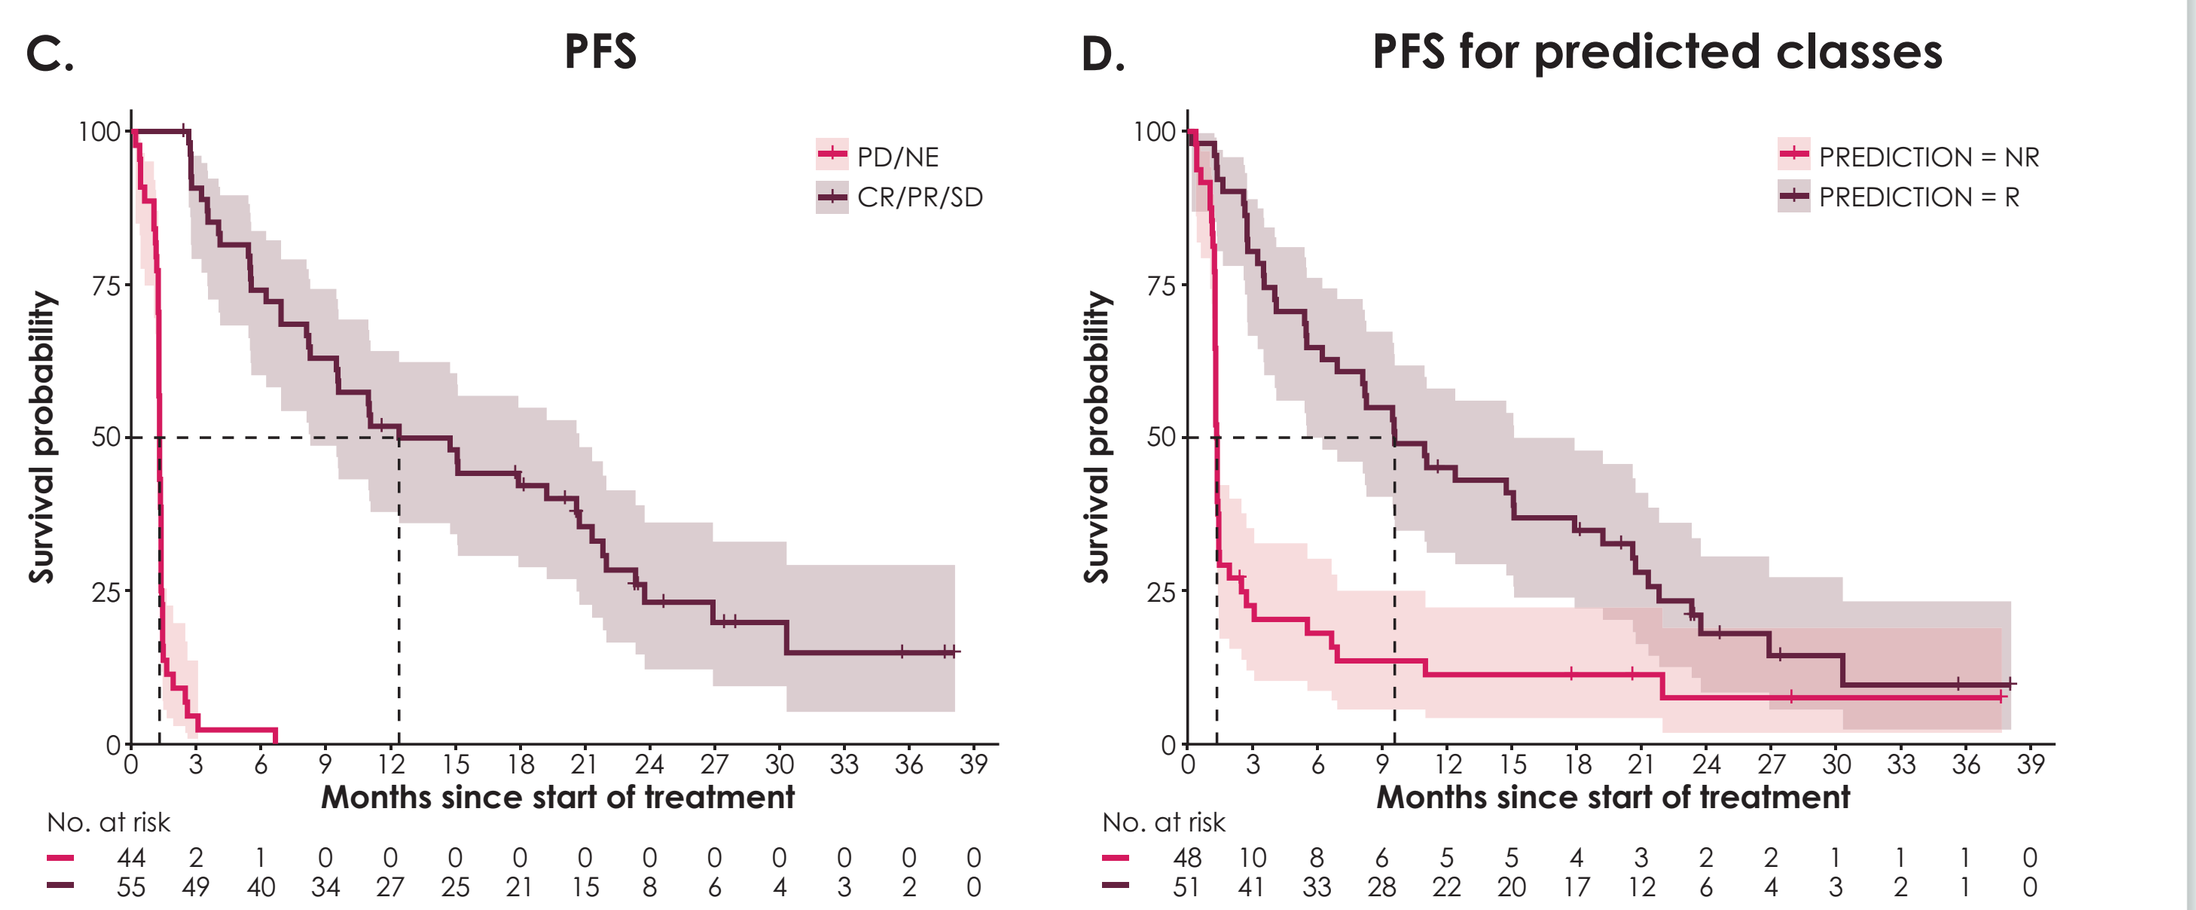
C.
PFS
PD/NE
CR/PR/SD
D.
PFS for predicted classes
PREDICTION = NR
PREDICTION = R
Survival probability
Months since start of treatment
No. at risk

C.

Response	n	Events	Median	0.95 LCI	0.95 UCI
NR (PD/NE)	44	44	1.31	1.28	1.38
R (CR/PR/SD)	55	41	12.39	8.28	20.73

D.

Response	n	Events	Median	0.95 LCI	0.95 UCI
NR (PD/NE)	48	43	1.365	1.28	1.45
R (CR/PR/SD)	51	42	9.59	5.52	15.11

LCI, lower confidence interval; **NA**, not available; **NR**, no response; **R**, response; **UCI**, upper confidence interval

Figure 4. Survival plots for PD-L1 subgroups

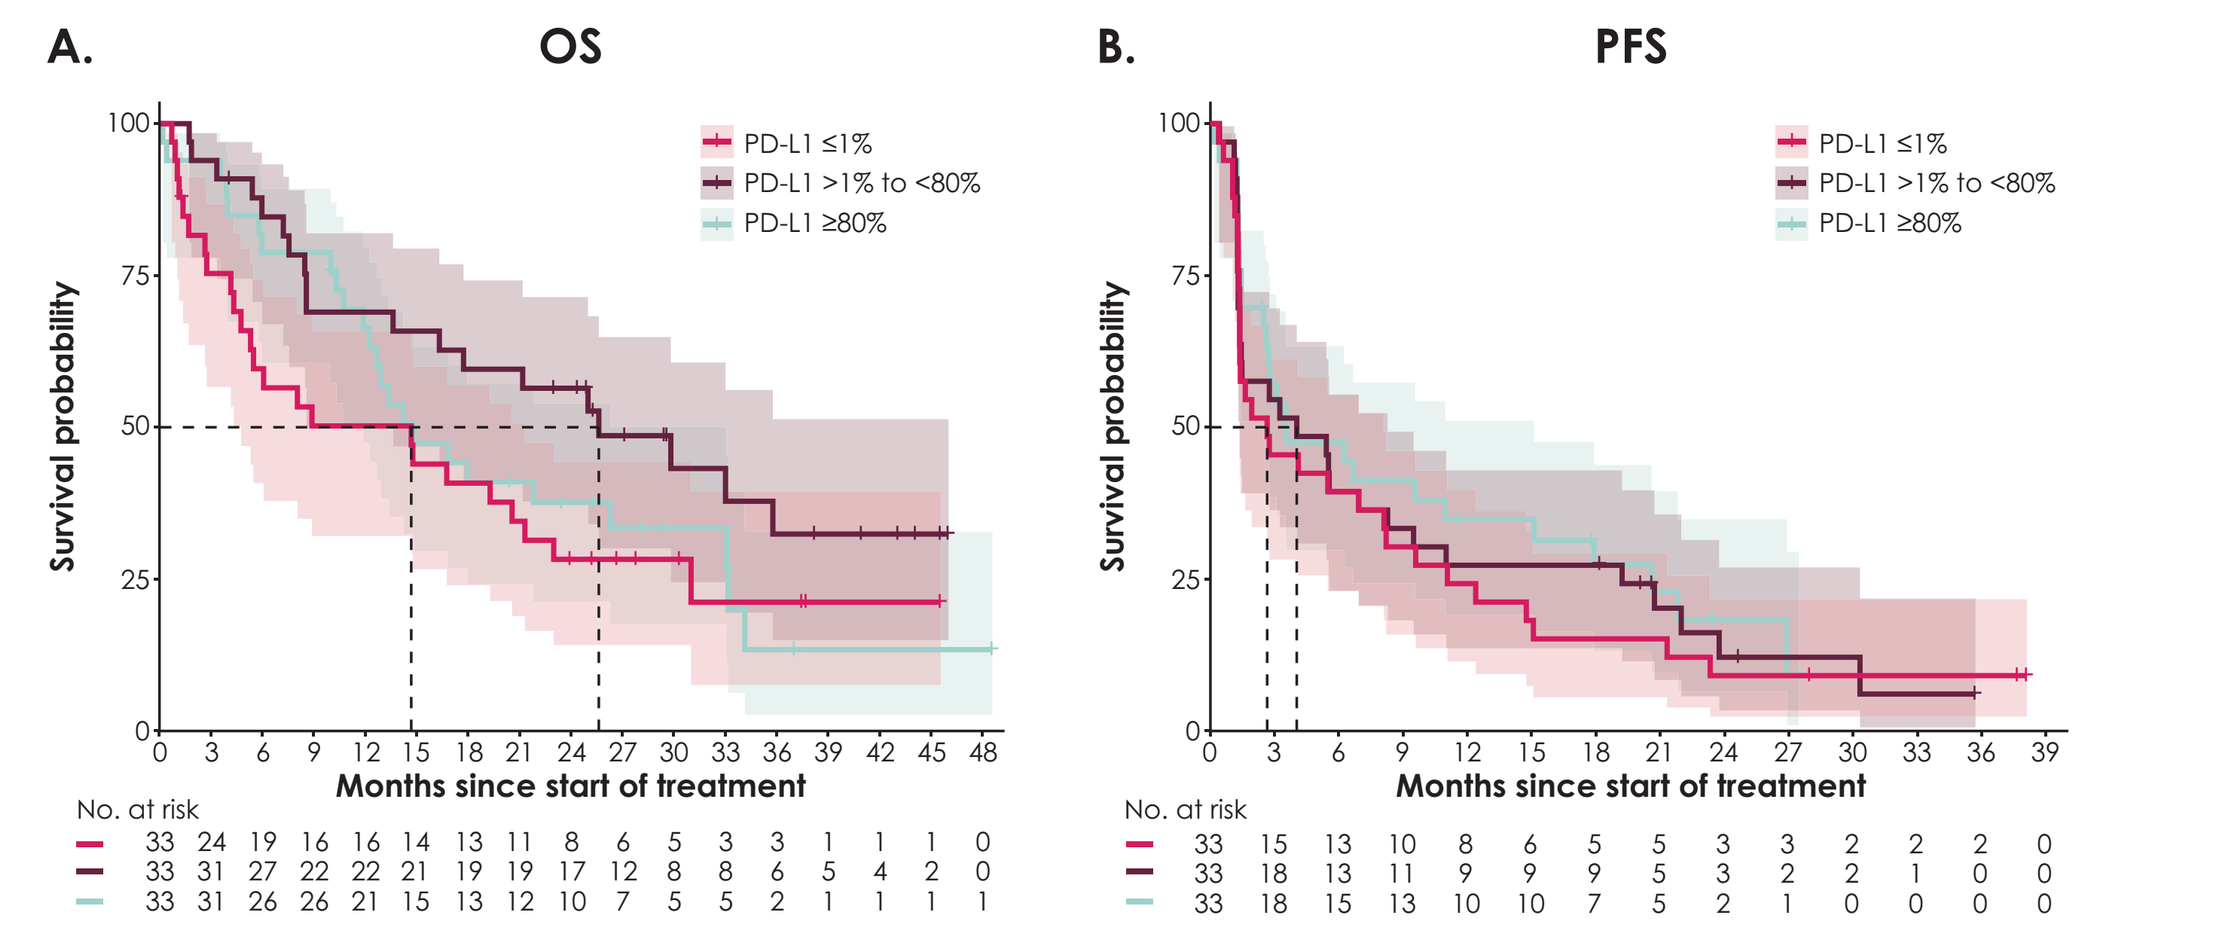
A.
OS
B.
PFS
PD-L1 ≤1%
PD-L1 >1% to <80%
PD-L1 ≥80%
Survival probability
Months since start of treatment
No. at risk

A.

PD-L1/OS	N	Events	Median	0.95 LCI	0.95 UCI
≤1%	33	24	14.7	4.76	21.3
>1% to <80%	33	19	25.6	8.57	NA
≥80%	33	24	14.8	11.89	33.1

B.

PD-L1/PFS	N	Events	Median	0.95 LCI	0.95 UCI
≤1%	33	30	2.66	1.38	8.11
>1% to <80%	33	29	4.04	1.38	8.28
≥80%	33	26	3.55	2.5	15.11

- The composite classifier had independent predictive power when assessed in the context of a multivariable Cox proportional hazards model (**Table 2**) and a multivariable logistic regression model (data not shown)

Table 2. Multivariable survival model suggesting independent power of classifier

Term	Level*	Exp (coef)	95% LCI	95% UCI	p value
(Intercept)		1.27	0.19	8.36	0.8
Prediction	(NR) R	45.07	12.86	212.99	0
Treatment line	(1L) 2L	0.93	0.26	3.29	0.92
Sex	(F) M	0.15	0.03	0.55	0.01
ECOG PS	(0) ≥1	0.54	0.15	1.89	0.35
Histology	(Adenocarcinoma) Squamous cell carcinoma	4.25	0.89	24.14	0.08
Histology	(Adenocarcinoma) Other	1.6	0.12	29.07	0.74
Tumor volume at baseline	Tumor volume at baseline	0.99	0.97	1	0.09
PD-L1 (1%)	(Nonevaluable) Negative	0.63	0.06	5.66	0.68
PD-L1 (1%)	(Nonevaluable) Positive	1.51	0.32	7.25	0.59

ECOG PS, Eastern Cooperative Oncology Group performance status
* Level in brackets suggests baseline category

- We also demonstrated that classifiers could be built with power to predict response across indications (**Figure 5A**) and combination treatment settings (**Figure 5B**)
- As observed in the avelumab and pembrolizumab 2L NSCLC analysis, the pembrolizumab monotherapy and combination classifiers had lower NPV, suggesting potential differences in non-responder profiles (**Figure 5**)

Figure 5. Application of monotherapy signatures to predict response across indications and for treatment combinations

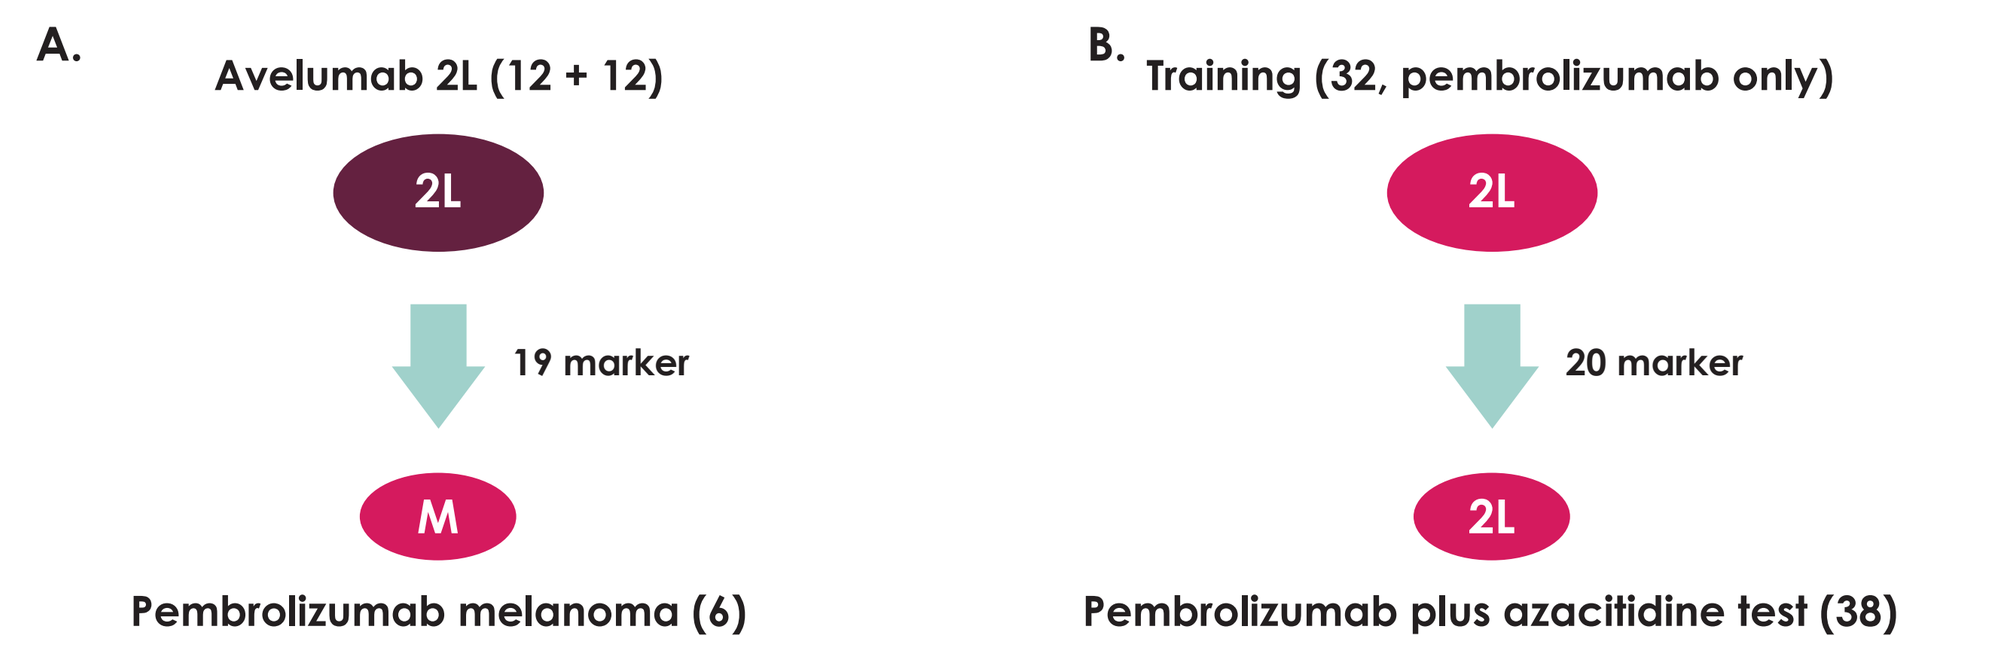
A.
Avelumab 2L (12 + 12)
2L
19 marker
M
Pembrolizumab melanoma (6)
B.
Training (32, pembrolizumab only)
2L
20 marker
2L
Pembrolizumab plus azacitidine test (38)

A. Performance on response

	NSCLC training	Pembrolizumab melanoma
Accuracy (95% CI)	0.95 (0.79-0.99)	1.0 (0.54-1.0)
PPV	1.0	1.0
NPV	0.92	1.0

B. Performance on response

	Training set	Pembrolizumab + azacitidine
Accuracy (95% CI)	0.97 (0.84-0.99)	0.71 (0.54-0.85)
PPV	1.0	0.72
NPV	0.86	0.67

CONCLUSIONS

- **Profiling patients who received avelumab or pembrolizumab with EpiSwitch demonstrated that:**
 - **It is possible to derive predictive signatures from baseline patient profiles that are applicable across agents with similar modalities, treatment lines, and indications**
 - **The biomarkers assayed using the EpiSwitch platform had high effect size because the training process requires a small number of samples to train**
 - **The signatures were small in size, provided good generalization on test sets (eg, PPV>0.7), and had independent predictive power**
- **Limitations**
 - **The single-arm design did not allow differentiation between the prognostic and predictive values of the classifiers**
- **Future work**
 - **Apply the classifier(s) to a blinded/independent test set that reflects real-world populations of a clinical trial and that also has a comparator arm**

REFERENCES

1. Tordini F, et al. Front Genet. 2016;7:194.
2. Barlesi F, et al. Lancet Oncol. 2018;19:1468-79.

ACKNOWLEDGMENTS

The authors would like to thank patients enrolled in the JAVELIN Solid Tumor trial for consenting to usage of samples for research purposes. This work was sponsored by Merck KGaA, Darmstadt, Germany, and EMD Serono (a business of Merck KGaA) and is part of an alliance between Merck KGaA, Darmstadt, Germany, and Pfizer Inc., New York, NY, USA.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SITC 2019 and the author of this poster.

For questions, please contact **parantu.shah@emdserono.com**.

GET POSTER PDF

Poster No. P143. Presented at SITC 2019; November 6-10, 2019; National Harbor, Maryland, USA.